Intraocular Pressure Reduction with NCX 470 versus Latanoprost In Previously Treated Versus Treatment-Naïve Patients

[1]Steven L. Mansberger, MD, MPH and [2]Doug Hubatsch, MSc., on behalf of the Mont Blanc Investigators

[1]Legacy Devers Eye Institute, [2]Nicox Ophthalmics, Inc.

Introduction

This was a post-doc analysis of the Mont Blanc Phase III clinical trial (NCT 04445519) evaluating the safety and efficacy of NCX470 0.1% versus latanoprost 0.005% in patients with open-angle glaucoma or ocular hypertension to determine if there was a difference in IOP response of patients previously treated with IOP-lowering medication versus treatment naïve.

Purpose

The purpose of this investigation was to compare intraocular pressure (IOP) reduction from baseline with NCX 470 0.1% QD versus latanoprost 0.005% QD in subjects with open-angle glaucoma (OAG) or ocular hypertension (OHT) who were and were not treatment-naïve at study entry.

Methods

- This was a post hoc analysis of the intent-to-treat population (N=661) of the MONT BLANC clinical trial.
- Eligibility criteria included unmedicated IOP $\geq$26 mmHg at 8AM, $\geq$24 mmHg at 10AM, and $\geq$22 mmHg at 4PM in the study eye.
- Previously treated subjects underwent washout prior to eligibility assessment.
- Subjects in the NCX 470 and latanoprost groups were divided into previously treated and treatment naïve groups for the analysis.
- Least-squares mean IOP reductions were calculated at 8AM and 4PM separately as well as combined (diurnal IOP: mean of 8AM and 4PM time points) at weeks 2 and 6 and month 3 from analysis of covariance models.

Results

Treatment Naïve

- In the treatment naïve subgroup (n=137 eyes), the difference in mean IOP reduction with NCX 470 and latanoprost numerically favored NCX 470 at 4 of 6 the time points (by 0.47-1.2 mmHg) and latanoprost at 2 of 6 time points (by 0.33 mmHg).
- The between-treatment difference in mean diurnal IOP favored NCX 470 numerically at all 3 visits (by 0.08-0.72 mmHg) in treatment naïve eyes.

Subjects Treatment Naïve at Screening

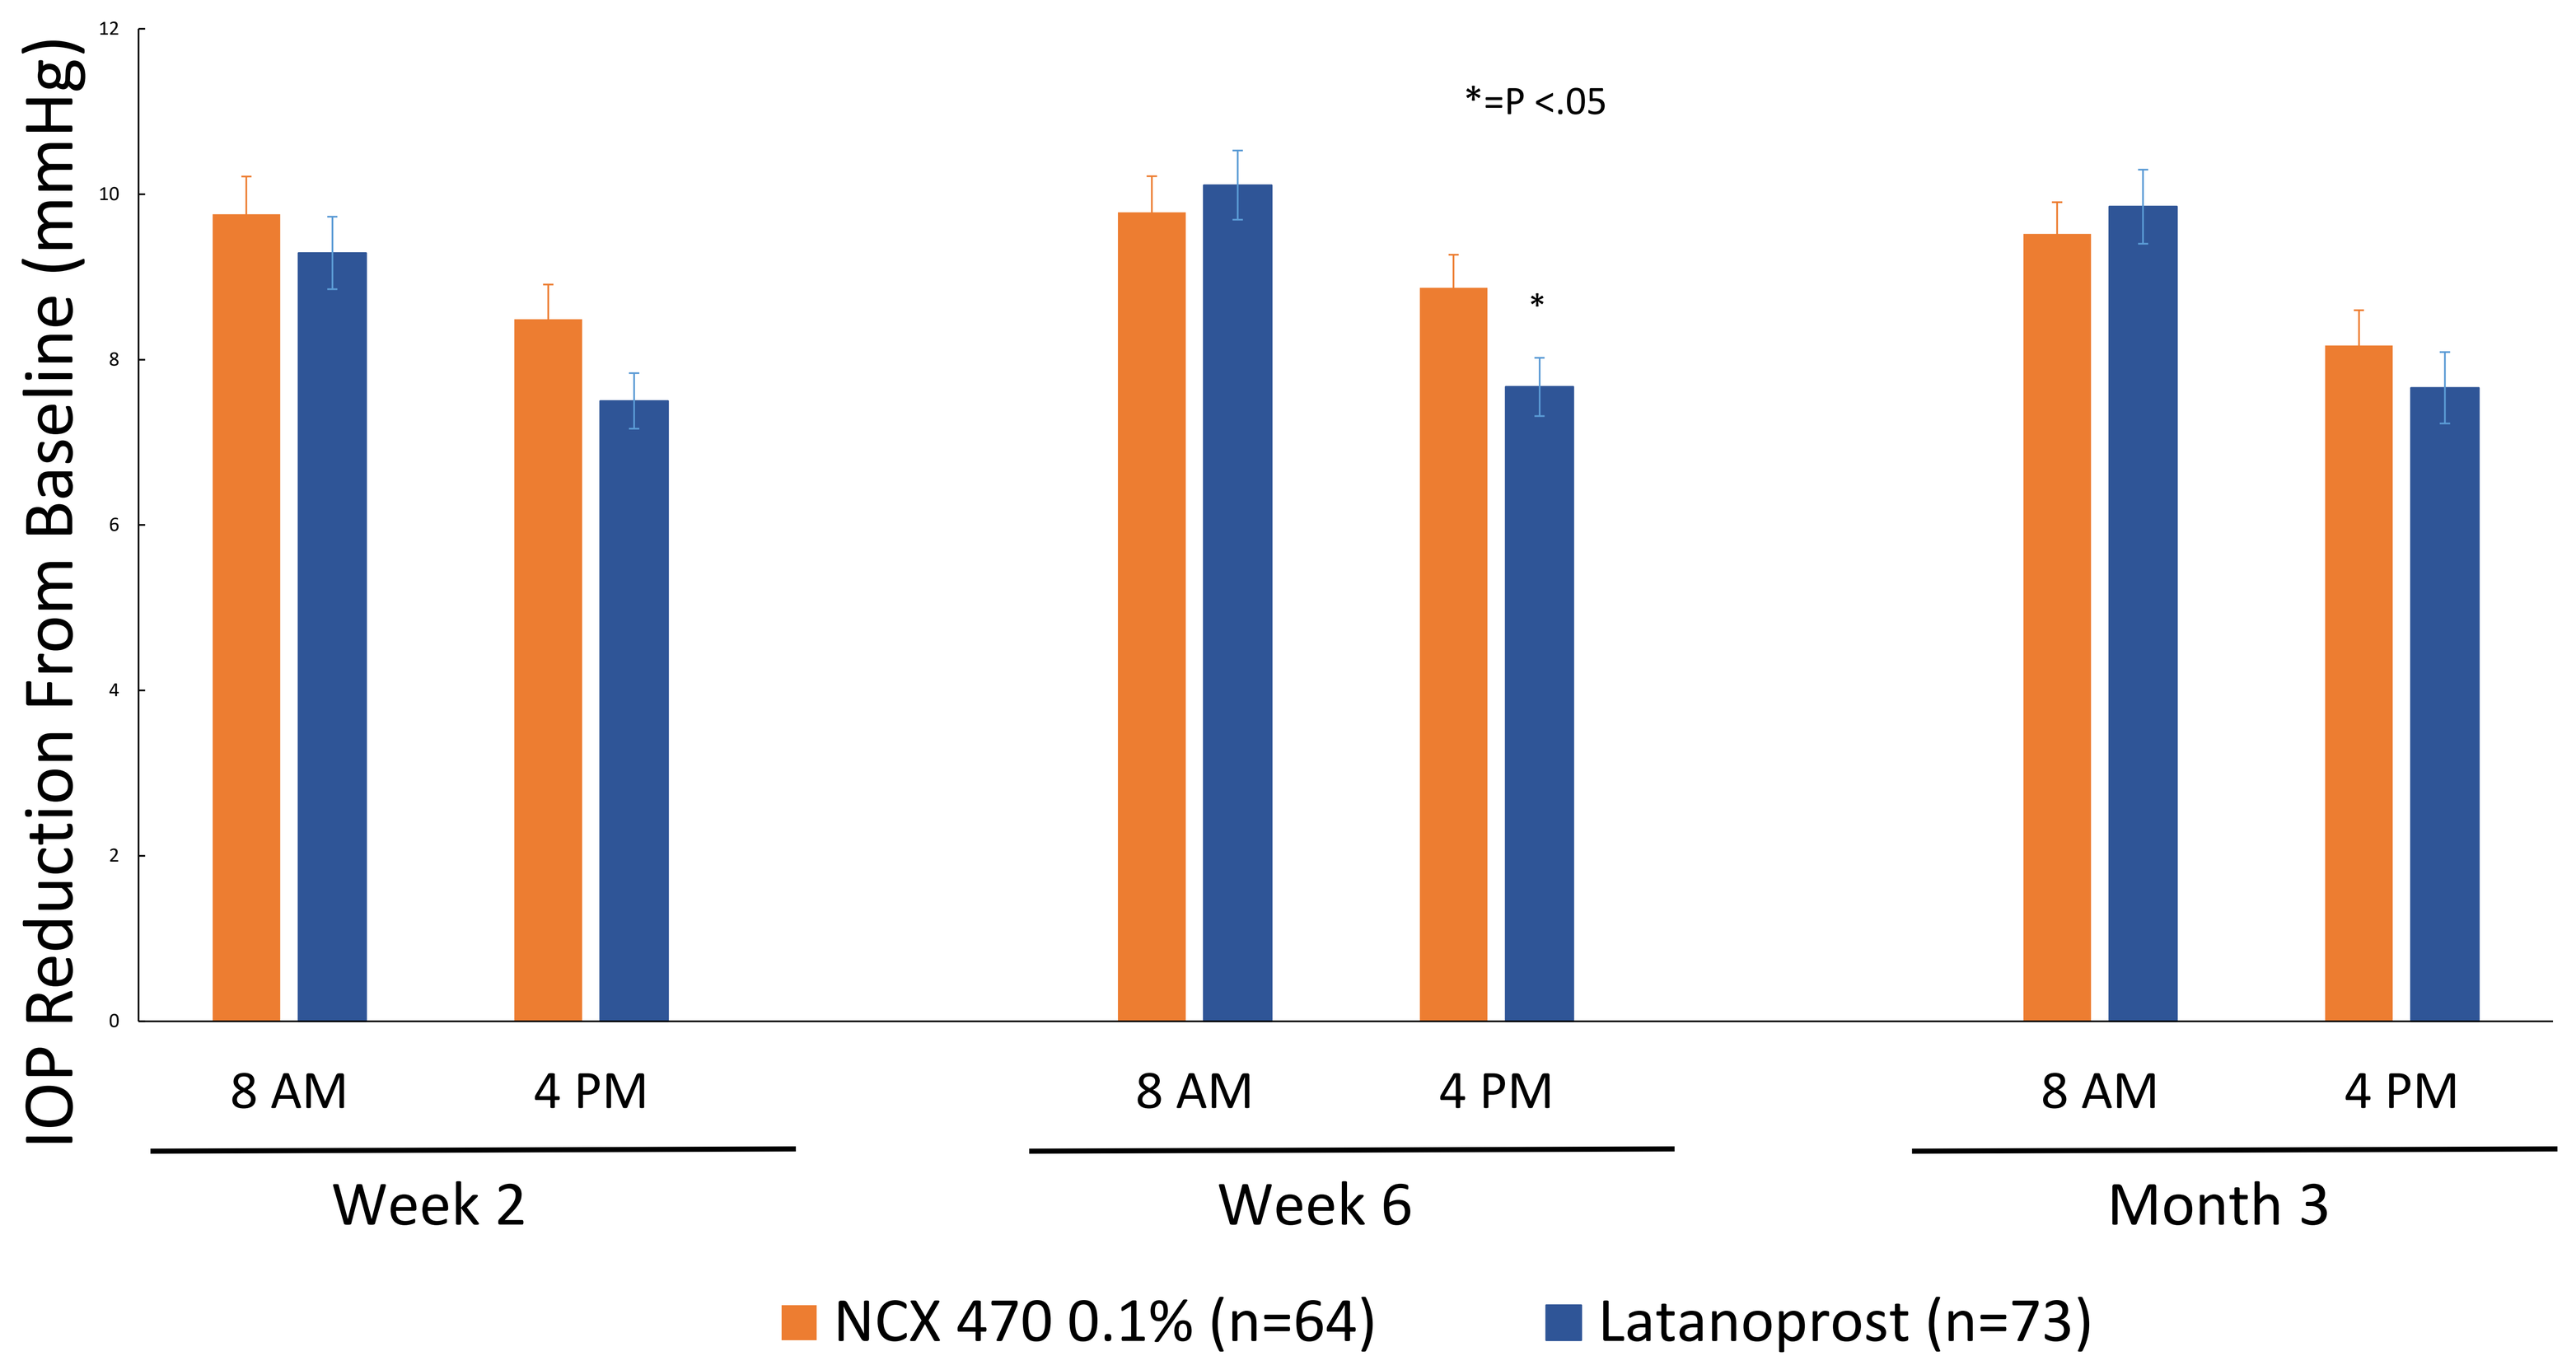
IOP Reduction From Baseline (mmHg)
0
2
4
6
8
10
12
*=P <.05
*
8 AM
4 PM
8 AM
4 PM
8 AM
4 PM
Week 2
Week 6
Month 3
NCX 470 0.1% (n=64)
Latanoprost (n=73)

Results

Previously Treated

- In the previously treated subgroup (n=524 eyes), the difference in mean IOP with NCX 470 and latanoprost favored NCX 470 numerically at all 6 time points (by 0.35-0.96 mmHg). The between-treatment difference in mean diurnal IOP favored NCX 470 numerically at all 3 visits (by 0.53-0.73 mmHg) in previously treated eyes.

Subjects Previously Treated with IOP-Lowering Medications at Screening

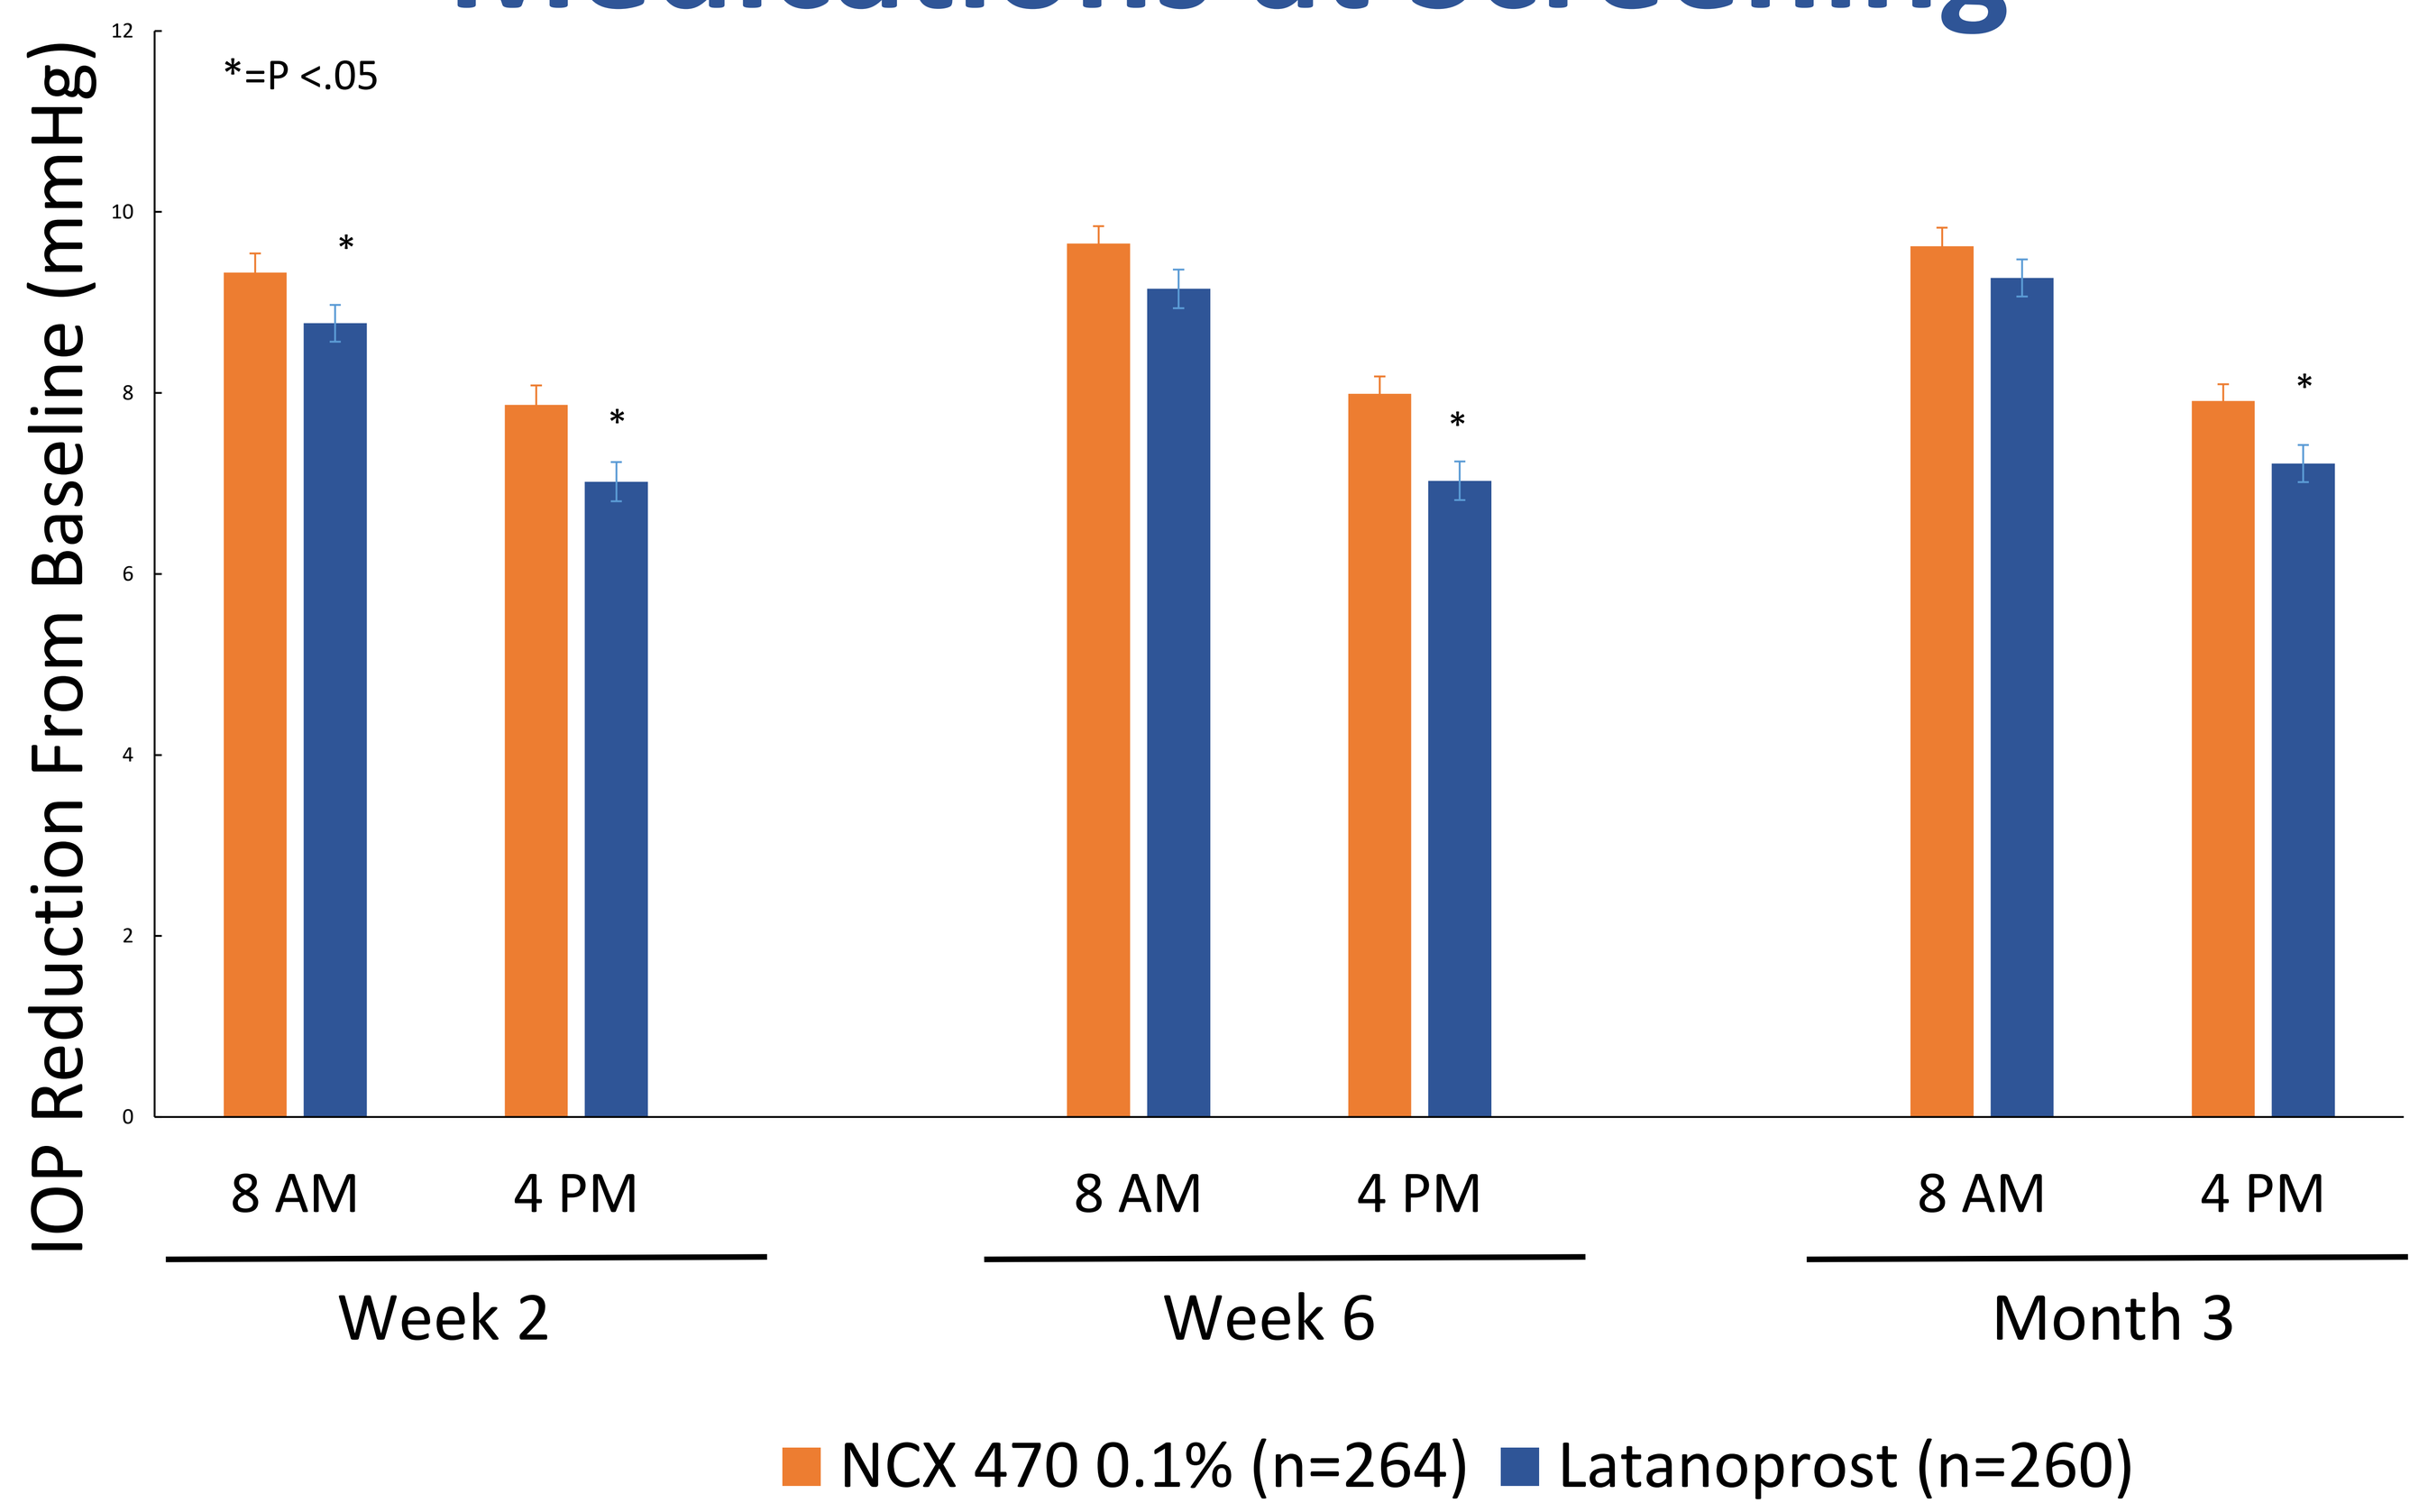
IOP Reduction From Baseline (mmHg)
0
2
4
6
8
10
12
*=P <.05
*
*
*
*
8 AM
4 PM
8 AM
4 PM
8 AM
4 PM
Week 2
Week 6
Month 3
NCX 470 0.1% (n=264)
Latanoprost (n=260)

Discussion

- Interestingly, previously treated subjects had numerically superior IOP with NCX 470 when compared to latanoprost across all time points but not by a clinically significant margin.

Conclusion

- Mean IOP reductions at 8AM and 4PM, as well as mean diurnal IOP (across the 3 study visits) had small differences between between NXC 470 and latanoprost in both treatment naïve eyes and previously treated eyes.
- Whether previously treated or treatment naïve-should not impact the selection of NCX 470 versus latanoprost in the treatment of OAG or OHT.

RESEARCH POSTER PRESENTATION DESIGN © 2022
www.PosterPresentations.com